A Systems Pharmacology Model of Anandamide Dynamics After FAAH Inhibitor Administration

Eugeniy Metlkin*(1), Natalia Bagrova(1), Oleg Demin(1), Neil Benson(2)

metelkin@insysbio.ru

(1) Institute for Systems Biology SPb, Moscow, Russia; (2) Pfizer Global Research and Development, Sandwich, UK

INTRODUCTION

Fatty acid amide hydrolase (FAAH) is an integral membrane enzyme hydrolyzing the anandamide and related amidated lipids. Previously it was shown preclinically that the inactivation of FAAH produces analgesic, anti-inflammatory, anxiolytic, and antidepressant effects indicating that FAAH may be a promising therapeutic target. This work describes a detailed mathematical model of anandamide and other ethanolamide kinetics that have been used for analysis of clinical data of PF-04457845 (Pfizer), a highly selective inhibitor of FAAH.

MODEL

The developed model includes the synthesis and hydrolysis of five major ethanolamides (AEA, OEA, PEA, LEA, SEA) in different tissues and organs as well as the processes of ethanolamide distribution. The detailed kinetic mechanism of each process was included into the model.

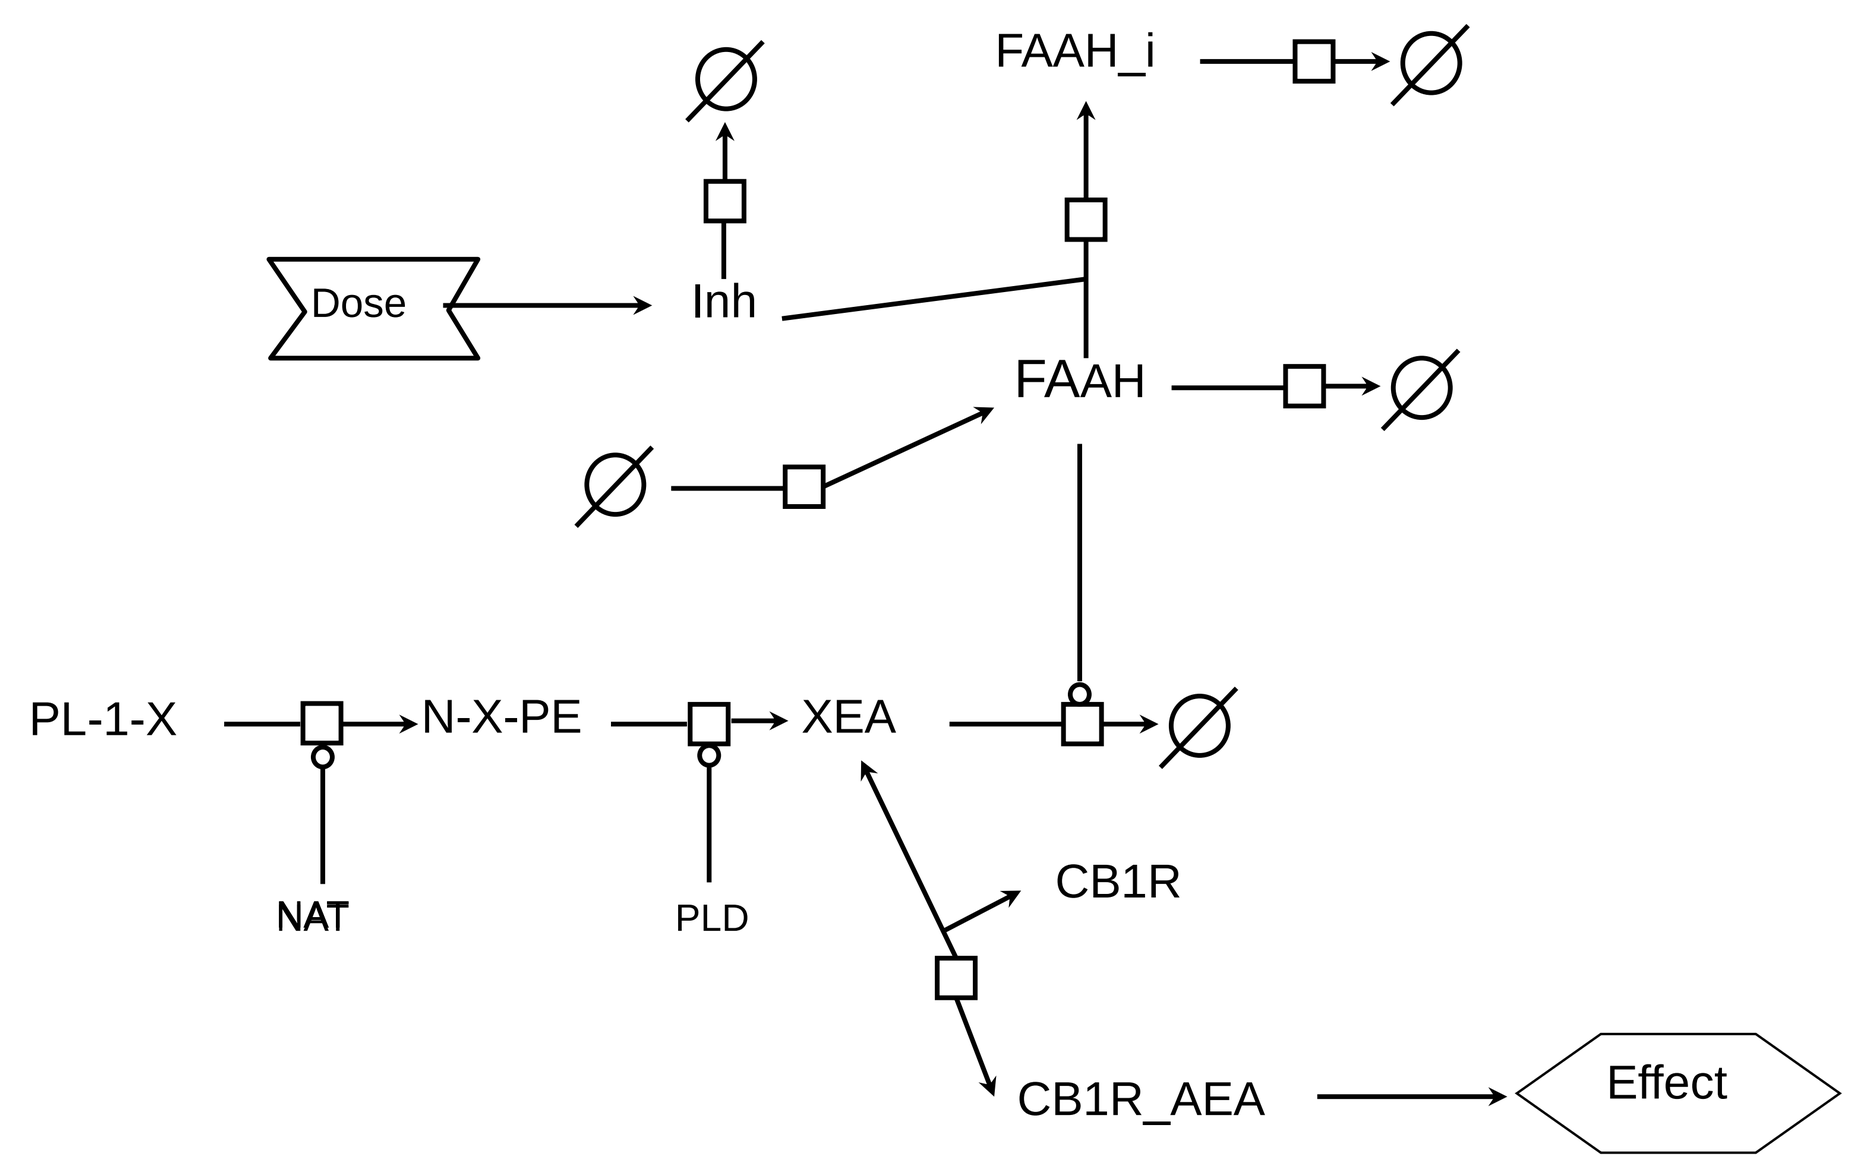
FAAH_i
Dose
Inh
FAAH
PL-1-X
N-X-PE
XEA
NAT
PLD
CB1R
CB1R_AEA
Effect

Fig.1. General scheme of fatty acid ethanolamide metabolism with irreversible FAAH inhibition (inh). X stands for one of five major amides: A, O, P, L, S.

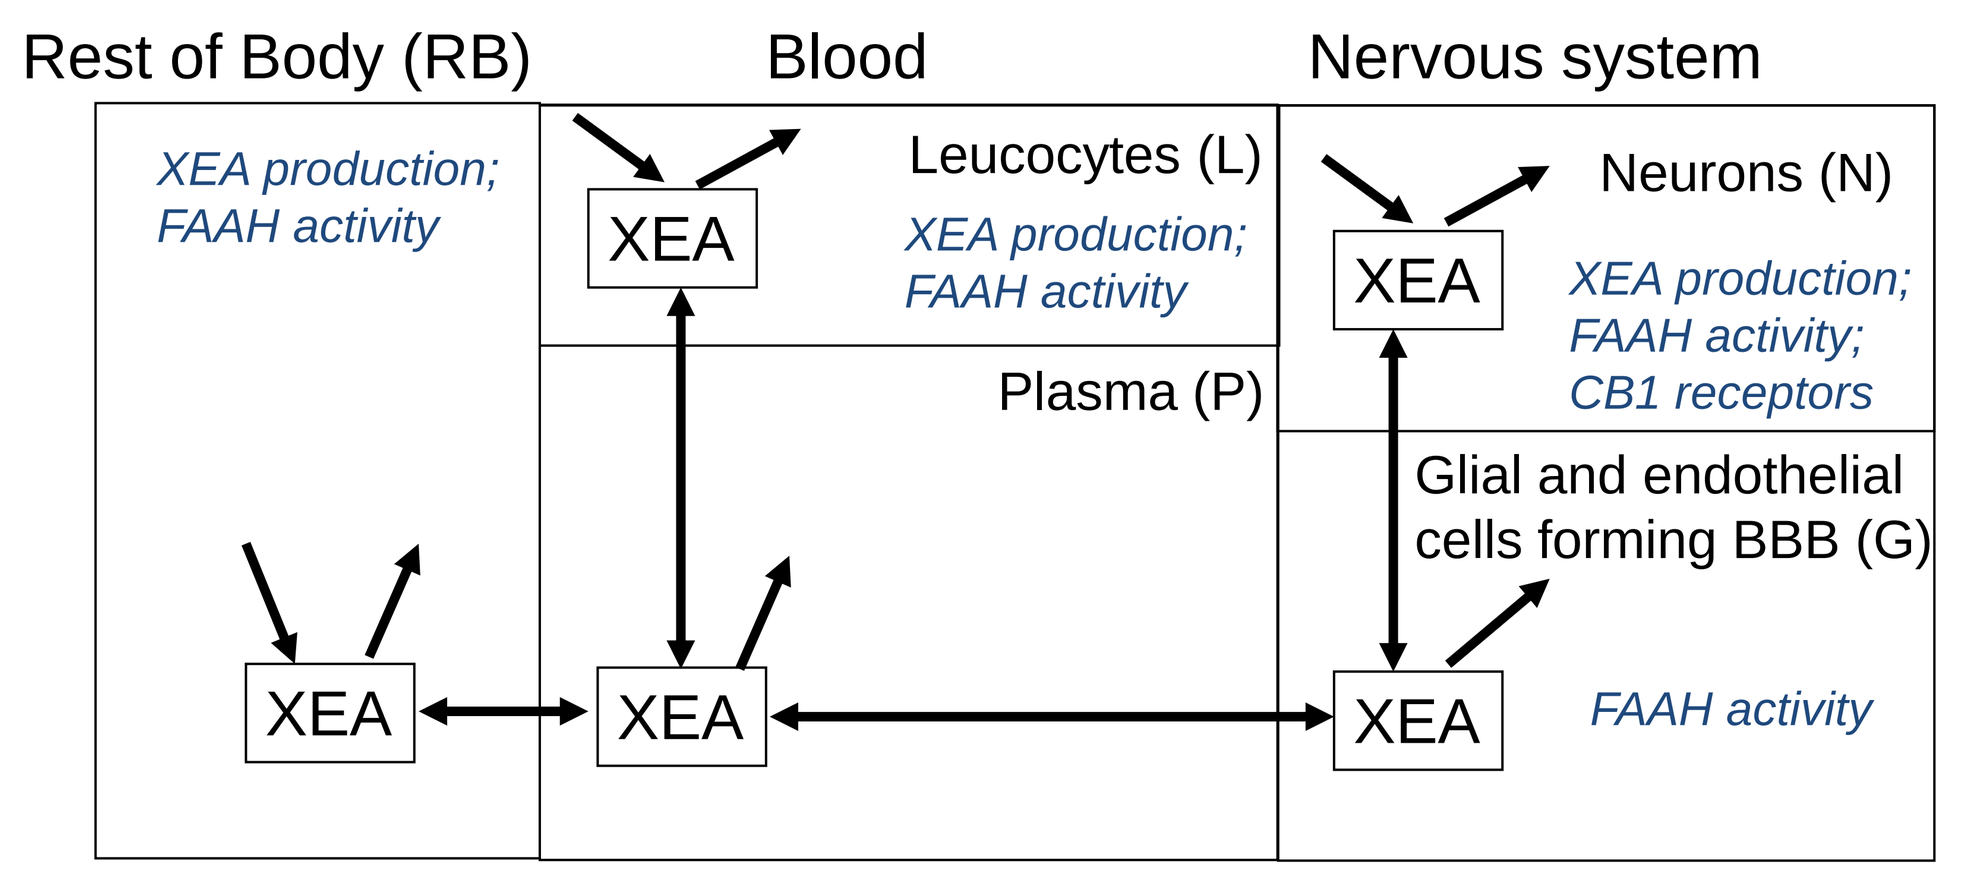
Rest of Body (RB)
Blood
Nervous system
XEA production; FAAH activity
Leucocytes (L)
XEA production; FAAH activity
Neurons (N)
XEA production; FAAH activity; CB1 receptors
Plasma (P)
Glial and endothelial cells forming BBB (G)
FAAH activity
XEA

Fig.2. Scheme of model compartments. Each metabolite: AEA, OEA, PEA, LEA or SEA was described as independent variable in each compartment. The PK of FAAH inhibitor and simple distribution were also described.

- ✓ The model of AEA, OEA, LEA, PEA, SEA synthesis was developed on the basis of NAT, PLD (fig.1) kinetic properties and data regarding the enzymes distribution in different tissues using the published data.
- ✓ The model of five fatty acid ethanolamide hydrolysis was developed on the basis of FAAH kinetic properties and known distribution of the enzyme in tissues.
- ✓ The percent of CB1 receptors bound with AEA was used to characterize the effect of FAAH inhibitor (PF-04457845). The binding of AEA to CB1 receptor was described on the basis of Kd measured for human[1].
- ✓ The distribution coefficients (KpT) for PF-04457845 and 5 ethanolamides were calculated using QSAR approach. A simple distribution model to describe the molecule transport between tissues was used (fig.2).
- ✓ A global model of ethanolamide methabolism was constructed. Some PK/PD parameters of the model were estimated on the basis of Goodness-of-Fit criterion using the results of clinical trial I.

REFERENCES

1) British Journal of Pharmacology (2007) 152, 583–593

VERIFICATION

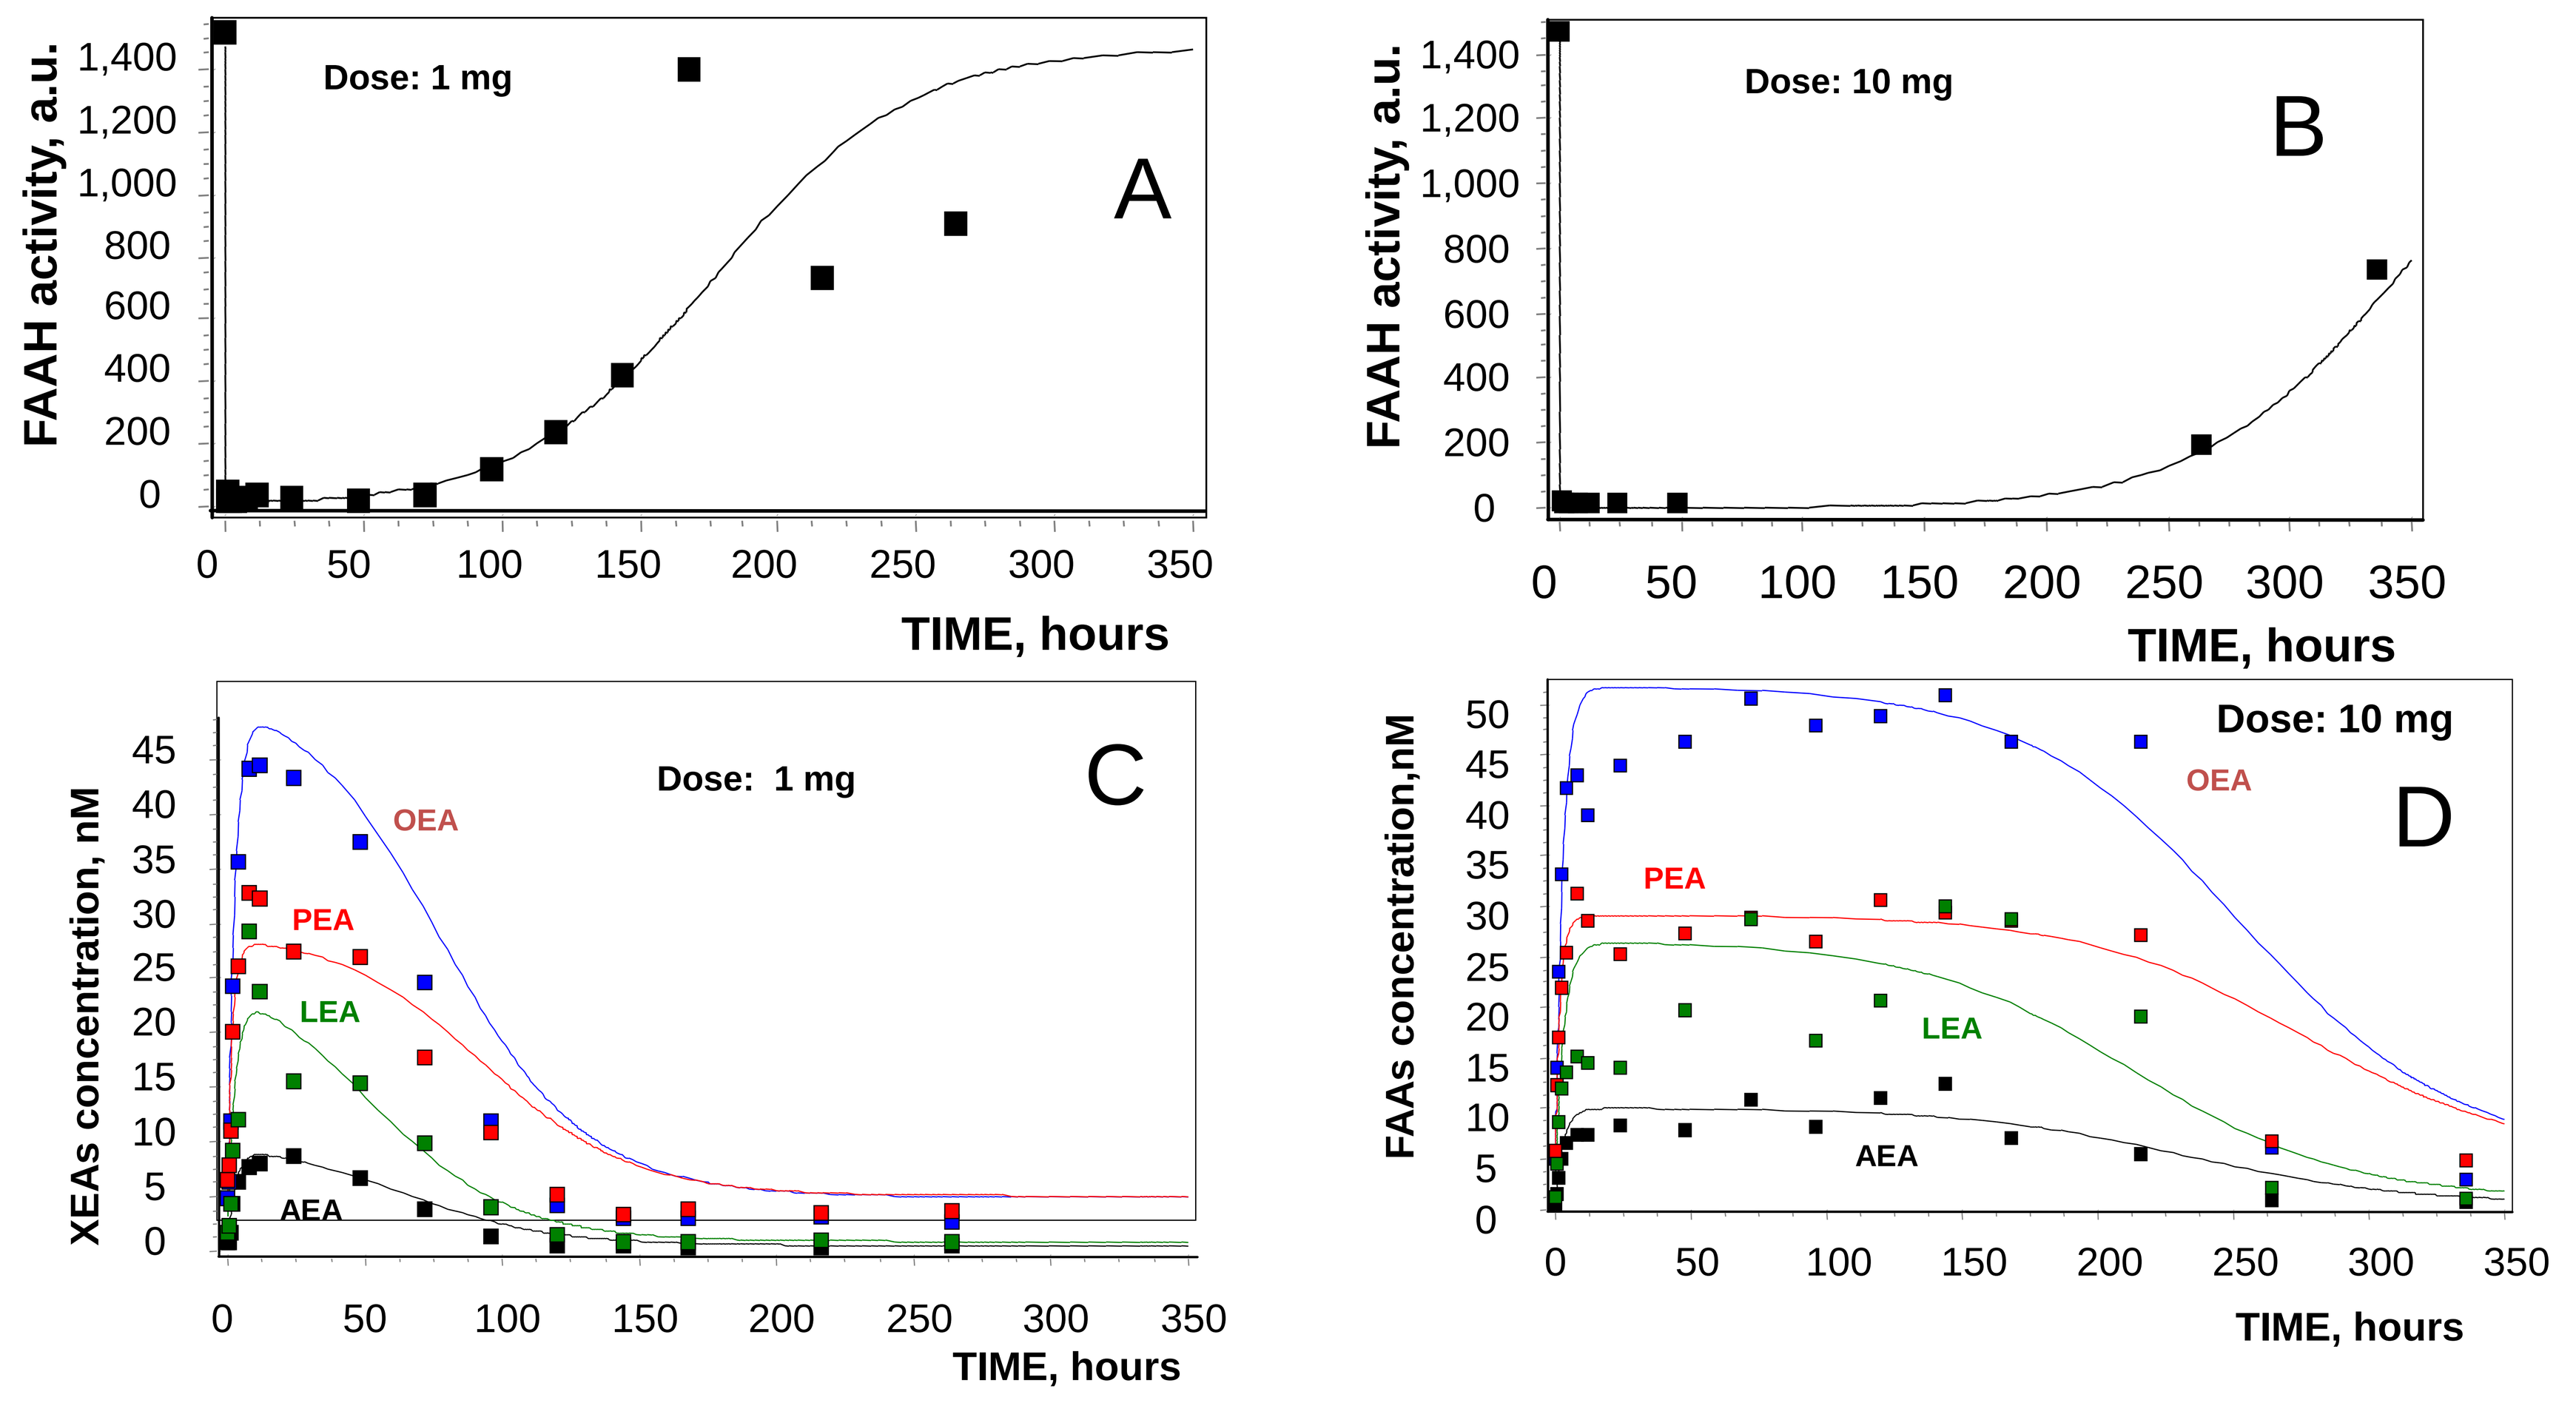
Dose: 1 mg
A
FAAH activity, a.u.
TIME, hours
Dose: 10 mg
B
C
Dose: 1 mg
XEAs concentration, nM
OEA
PEA
LEA
AEA
D
Dose: 10 mg
FAAs concentration,nM

Fig.3. Example of dataset used for parameter estimation and fitted theoretical curves: A),B) FAAH activity in blood at different doses; C), D) ethanolamide concentration in blood plasma at different doses.

RESULTS

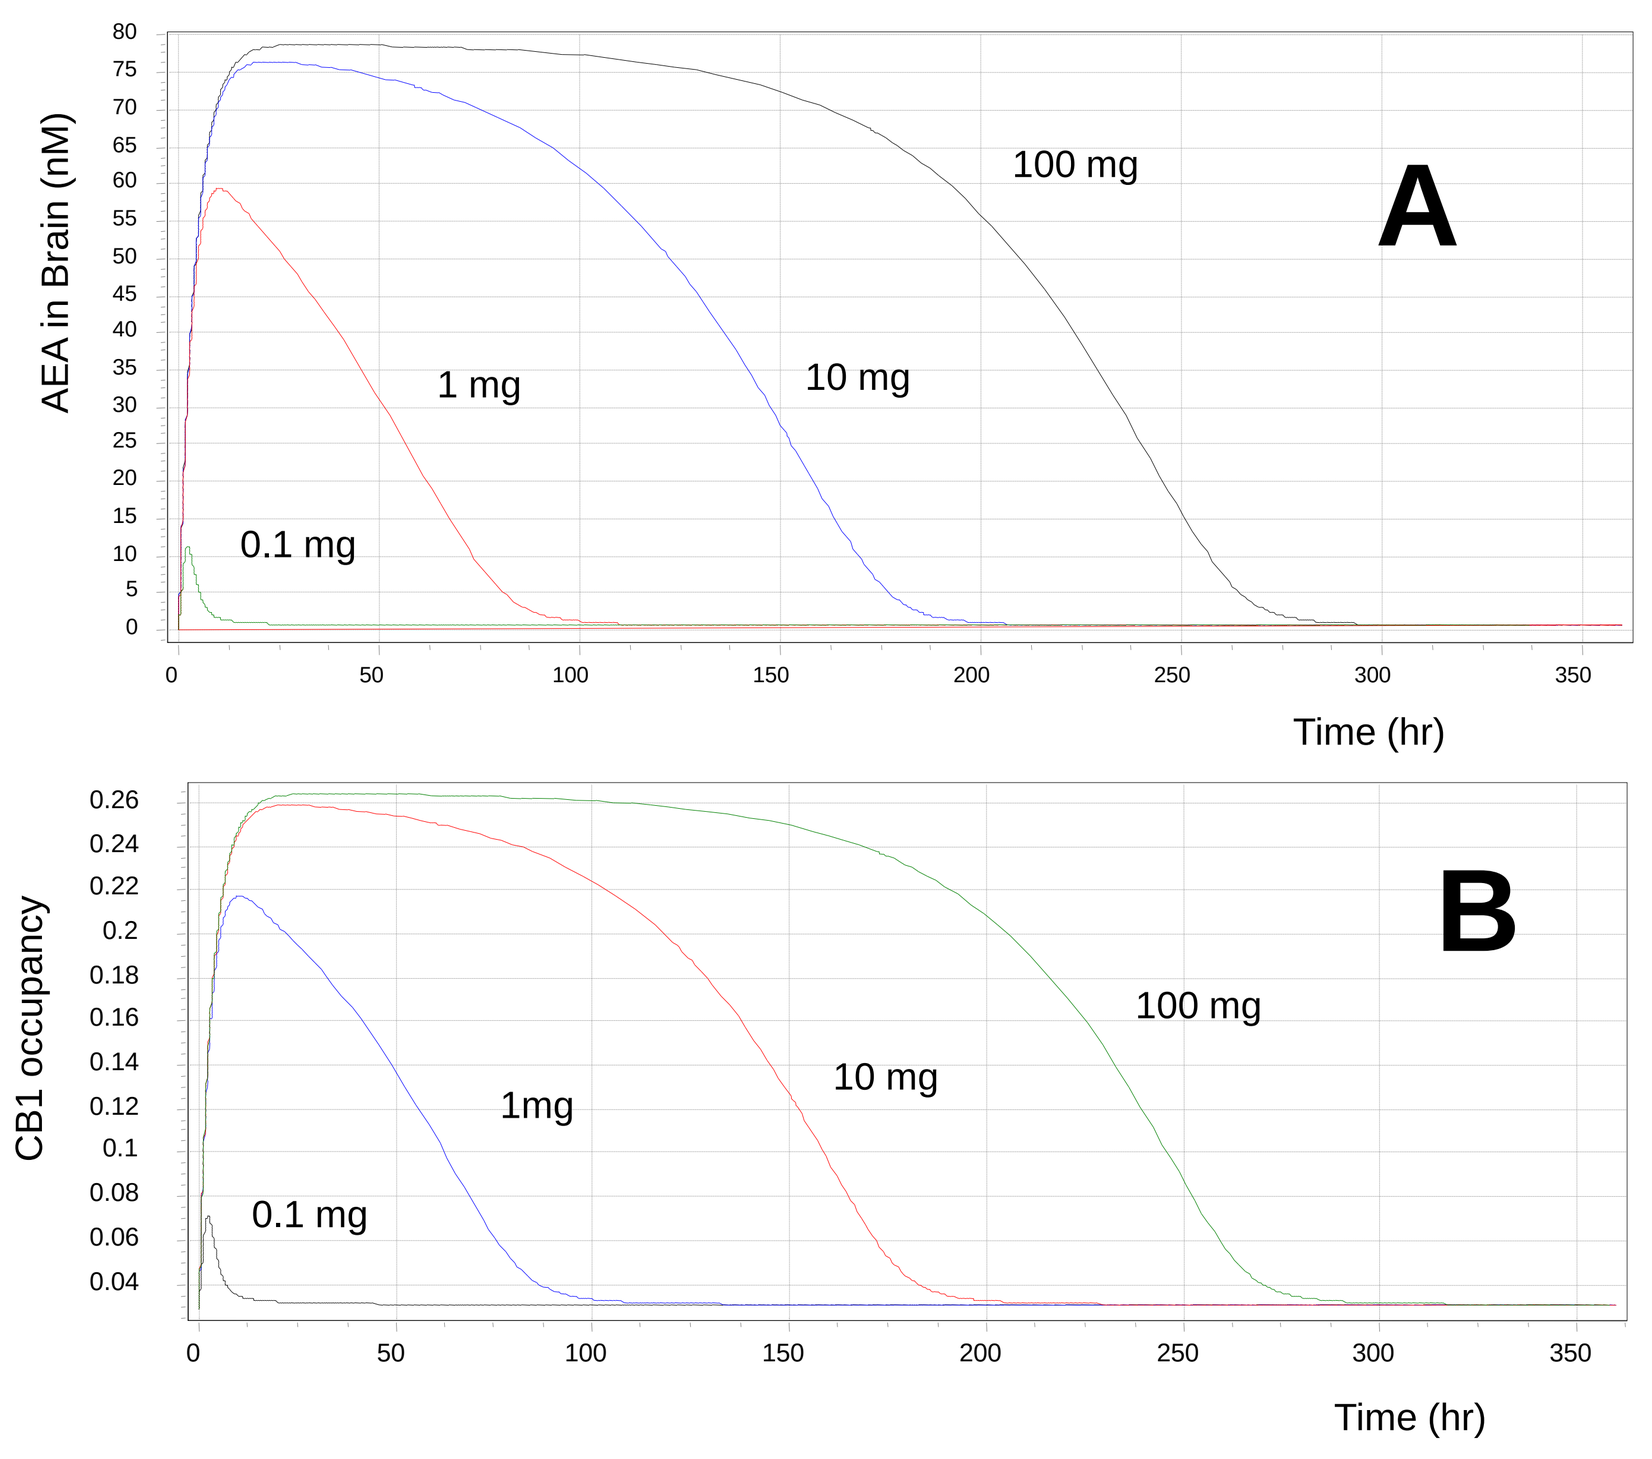
A
AEA in Brain (nM)
100 mg
10 mg
1 mg
0.1 mg
Time (hr)
B
CB1 occupancy
1mg

Fig. 4. Model based predicted values after administration of FAAH inhibitor for different single doses: A) average anandamide concentration in brain; B) CB1 receptor in brain, binded/total ratio.

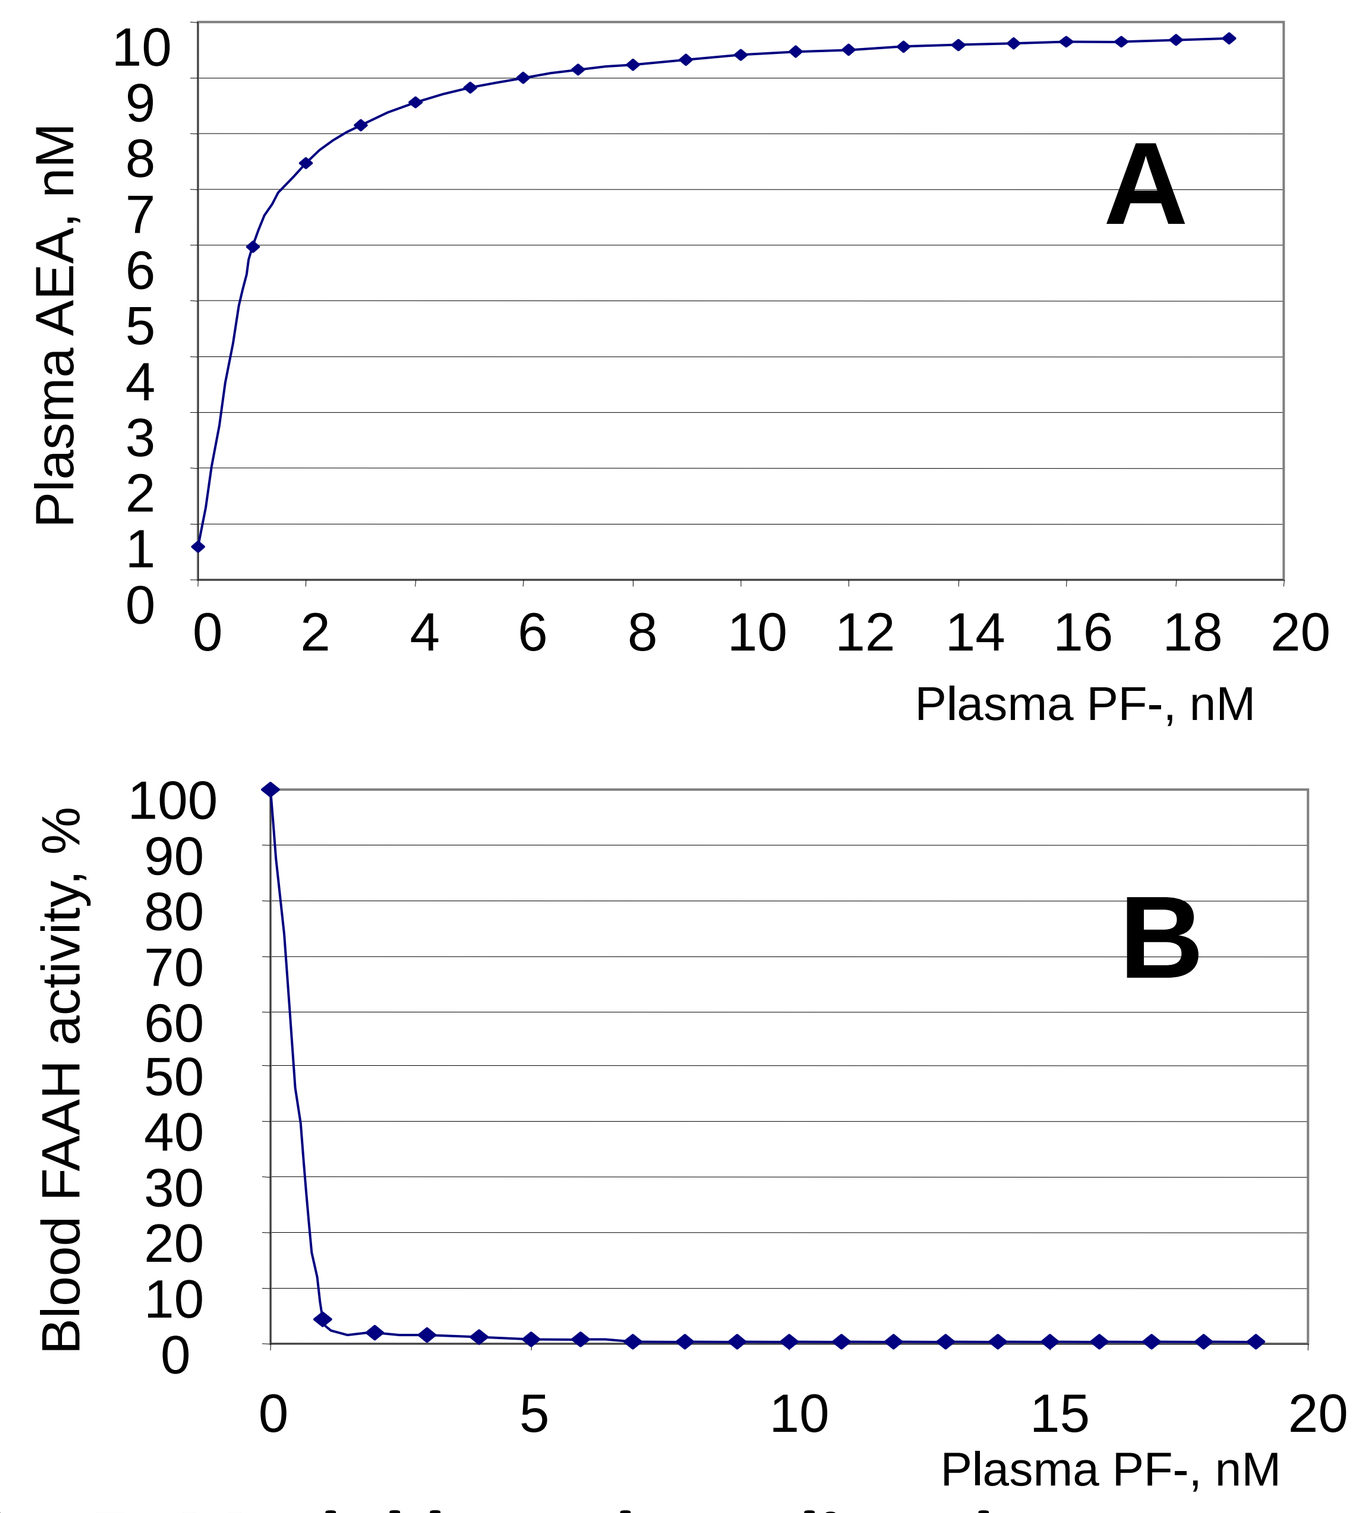
A
Plasma AEA, nM
Plasma PF-, nM
B
Blood FAAH activity, %

Fig. 5. Model based predicted concentration/effect relationships: A) anandamide concentration in blood plasma; B) FAAH activity in blood.

CONCLUSIONS

- ✓ The model of AEA metabolism was developed. The model simulate numerically different treatment regimes and effectiveness of FAAH inhibitors on the basis its PK and binding properties.
- ✓ The occupancy of CB1 receptor changes from 3% to 26% after drug (PF-04457845) administration with high doses.
- ✓ The model provides the basis for optimal dose and regime predictions of FAAH inhibitors for anti-pain treatment.
- ✓ The high rate of AEA additional degradation limits the process of AEA increase even after total inhibition of FAAH.

ACKNOWLEDGMENTS

The authors would like to thank Dr Piet H. Van Der Graaf for helpful discussions.

CONTACTS

For more information regarding our investigations, please visit our web site at www.insysbio.ru